Long-term overall survival and progression-free survival in idiopathic pulmonary fibrosis treated by pirfenidone or nintedanib or their switch: real-world data from the EMPIRE registry

Martina Vašáková,[1] Martina Šterclová,[1] Nesrin Mogulkoc,[2] Katarzyna Lewandowska,[3] Veronika Müller,[4] Marta Hájková,[5] Dragana Jovanovic,[6] Jasna Tekavec-Trkanjec,[7] Mordechai Kramer,[8] Michael Studnicka,[9] Natalia Stoeva,[10] Simona Littnerová,[11] Karel Hejduk,[11] Ladislav Dušek[11]

[1]Department of Respiratory Diseases of the First Faculty of Medicine Charles University, Thomayer Hospital, Prague, Czech Republic; [2]Department of Pulmonary Medicine, Ege University Medical School, Izmir, Turkey; [3]Department of Pulmonary Diseases, Institute of Tuberculosis and Lung Diseases, Warsaw, Poland; [4]Department of Pulmonology, Semmelweis University, Budapest, Hungary; [5]Clinic of Pneumology and Phthisiology, University Hospital Bratislava, Bratislava, Slovakia; [6]University Hospital of Pulmonology, Clinical Center of Serbia, Belgrade, Serbia; [7]Pulmonary Department, University Hospital Dubrava, Zagreb, Croatia; [8]Institute of Pulmonary Medicine, Rabin Medical Center, Petah Tikva, Israel; [9]Department of Pneumology, Paracelsus Medical University, SALK, Salzburg, Austria; [10]Pulmonary Department, Acibadem City Clinic Tokuda Hospital, Sofia, Bulgaria; [11]Institute of Biostatistics and Analyses of the Faculty of Medicine, Masaryk University, Brno, Czech Republic

http://bi_poster.staging.n241.com/22021888_Vasakova.pdf

PA4720

Introduction

- Antifibrotic treatment with either nintedanib or pirfenidone can slow down the progression of IPF but cannot cure it.[1–3]
- Evidence for the efficacy of antifibrotic treatment comes mostly from randomised clinical trials.[4–7] The long-term effect of antifibrotic treatment on the clinical course of IPF in the real world is not fully understood.

Aims

- We aimed to compare OS and PFS in patients from the European Multipartner IPF registry (EMPIRE; http://empire.registry.cz/) treated with either pirfenidone or nintedanib, or who switched from nintedanib to pirfenidone or vice versa, or who received other or no treatment.

Methods

- In this retrospective study, patients with IPF included in EMPIRE were diagnosed according to the ATS/ERS consensus classification.
- Patients who switched antifibrotic treatment were divided into groups based on reason for discontinuation of first therapy:
 - treatment switch due to AEs
 - treatment switch due to lack of treatment effect (defined by health insurance company rules or physician opinion).

OS and PFS

- Kaplan–Meier methodology was used to estimate median OS and PFS. OS and PFS were calculated from entry into the registry.
 - There was left truncation arising from the delay between diagnosis and enrolment into the registry by placing patients in the risk set only from time of enrolment.
- Progression was defined as death, transplantation or decline in FVC >10% or TL_{CO} >15% compared with previous values.

Results

Patient Characteristics

- A total of 2,745 patients with IPF from the EMPIRE registry were included and assigned to groups on the basis of antifibrotic treatment received (Table 1). Of these, 29.5% were female and 70.5% were male. Mean age at diagnosis was 66.5 years.
- For patients who switched between treatments, the median duration of first therapy was 8.05 months and median time to starting the second therapy was 0.25 months.
- There was no significant difference in baseline characteristics between patients who switched treatment due to AEs and those who switched due to lack of treatment effect.

Table 1. Patient characteristics

	Pirfenidone (n=821)	Nintedanib (n=637)	Treatment switch (n=148)	Other treatment (n=1,139)	P value[a]
Gender, n (%)					
Female	193 (23.5)[b]	204 (32.0)[b]	45 (30.4)[b]	367 (32.2)[b]	<0.001
Male	628 (76.5)	433 (68.0)	103 (69.6)	772 (67.8)	
Mean age at diagnosis, years (95% CI)	66 (65.6, 66.9)[b]	66 (65.1, 66.6)[b]	66 (64.9, 67.4)[b]	68 (67.4, 68.7)[b]	<0.001
Mean duration of symptoms at diagnosis, months (95% CI)[c]	12 (9.9, 14.0)[b]	12 (10.3, 14.4)[b]	13 (10.8, 15.1)[b]	10 (8.2, 12.2)[b]	0.003
HRCT[d], n (%)					
UIP pattern	621 (75.7)[b]	439 (69.0)[b]	116 (78.4)[b]	759 (66.8)[b]	<0.001
UIP possible	171 (20.9)	187 (29.4)	30 (20.3)	334 (29.4)	
Inconsistent with UIP	28 (3.4)	10 (1.6)	2 (1.4)	44 (3.9)	
Mean lung function at start of treatment, % (95% CI)[e]					
FVC % predicted[f]	72 (70.0, 73.2)[b]	79 (76.8, 80.6)[b]	74 (71.1, 76.5)[b]	76 (72.9, 78.2)[b]	<0.001
TL_{CO} % predicted[g]	45 (42.8, 46.7)[b]	47 (45.1, 48.8)[b]	50 (47.4, 52.8)[b]	42 (38.8, 44.4)[b]	<0.001

[a]Statistical difference was evaluated by Chi-squared test for categorical variables and ANOVA test for continuous variables. [b]Homogeneous group assessed by post hoc test. [c]Duration of symptoms at diagnosis (months) has log-normal distribution and was described by geometric mean (95% CI). [d]Data missing for four patients. [e](±3 months). [f]Data missing for 1,366 patients. [g]Data missing for 1,397 patients.

Survival

- Median OS was longer in the nintedanib group and in patients who switched treatment, compared with patients on pirfenidone (P=0.003) or those who received other treatment (P<0.001), respectively (Figure 1A). OS rates at 1, 5 and 10 years were:
 - nintedanib vs pirfenidone: 0.912 vs 0.872, 0.430 vs 0.311 and 0.197 vs 0.105, respectively
 - treatment switch vs other treatment: 0.961 vs 0.688, 0.540 vs 0.138 and 0.347 vs 0.023, respectively.
- Median PFS was similar in the nintedanib group vs the pirfenidone group (P=0.200) and longer in patients who switched treatment vs those who received other treatment (P<0.001) (Figure 1B). PFS rates at 1, 5 and 10 years were:
 - nintedanib vs pirfenidone: 0.669 vs 0.673, 0.232 vs 0.173 and 0.055 vs 0.040, respectively
 - treatment switch vs other treatment: 0.789 vs 0.617, 0.333 vs 0.094 and 0.084 vs 0.008, respectively.
- Patients who switched treatment due to AEs had a numerically longer median OS than those who switched due to lack of treatment effect (P=0.395) (Figure 2A).
 - OS rates were similar in patients who switched treatment due to AEs vs those who switched due to lack of treatment effect (1 year: 0.969 vs 0.915; 2 years: 0.752 vs 0.810, respectively).
 - The 5-year OS rate was numerically higher in patients who switched treatment due to AEs (0.561) compared with those who switched due to treatment effect (0.126).
- There was no significant difference in PFS rate for patients who switched treatment due to AEs vs those who switched due to lack of treatment effect (P=0.898) and PFS rates were 0.722 vs 0.783 (1 year), 0.509 vs 0.592 (2 years) and 0.351 vs 0.102 (5 years), respectively.
 - Similar to the OS results, PFS diverged over time.

Lung Function

- Comparison of lung function between pirfenidone-treated and nintedanib-treated patients showed that similar proportions had FVC decline >10%, decline <10% or stable/increase in FVC at 6 (±2) months from diagnosis (data not shown).

Figure 1. Long-term survival

A. OS **B. PFS**

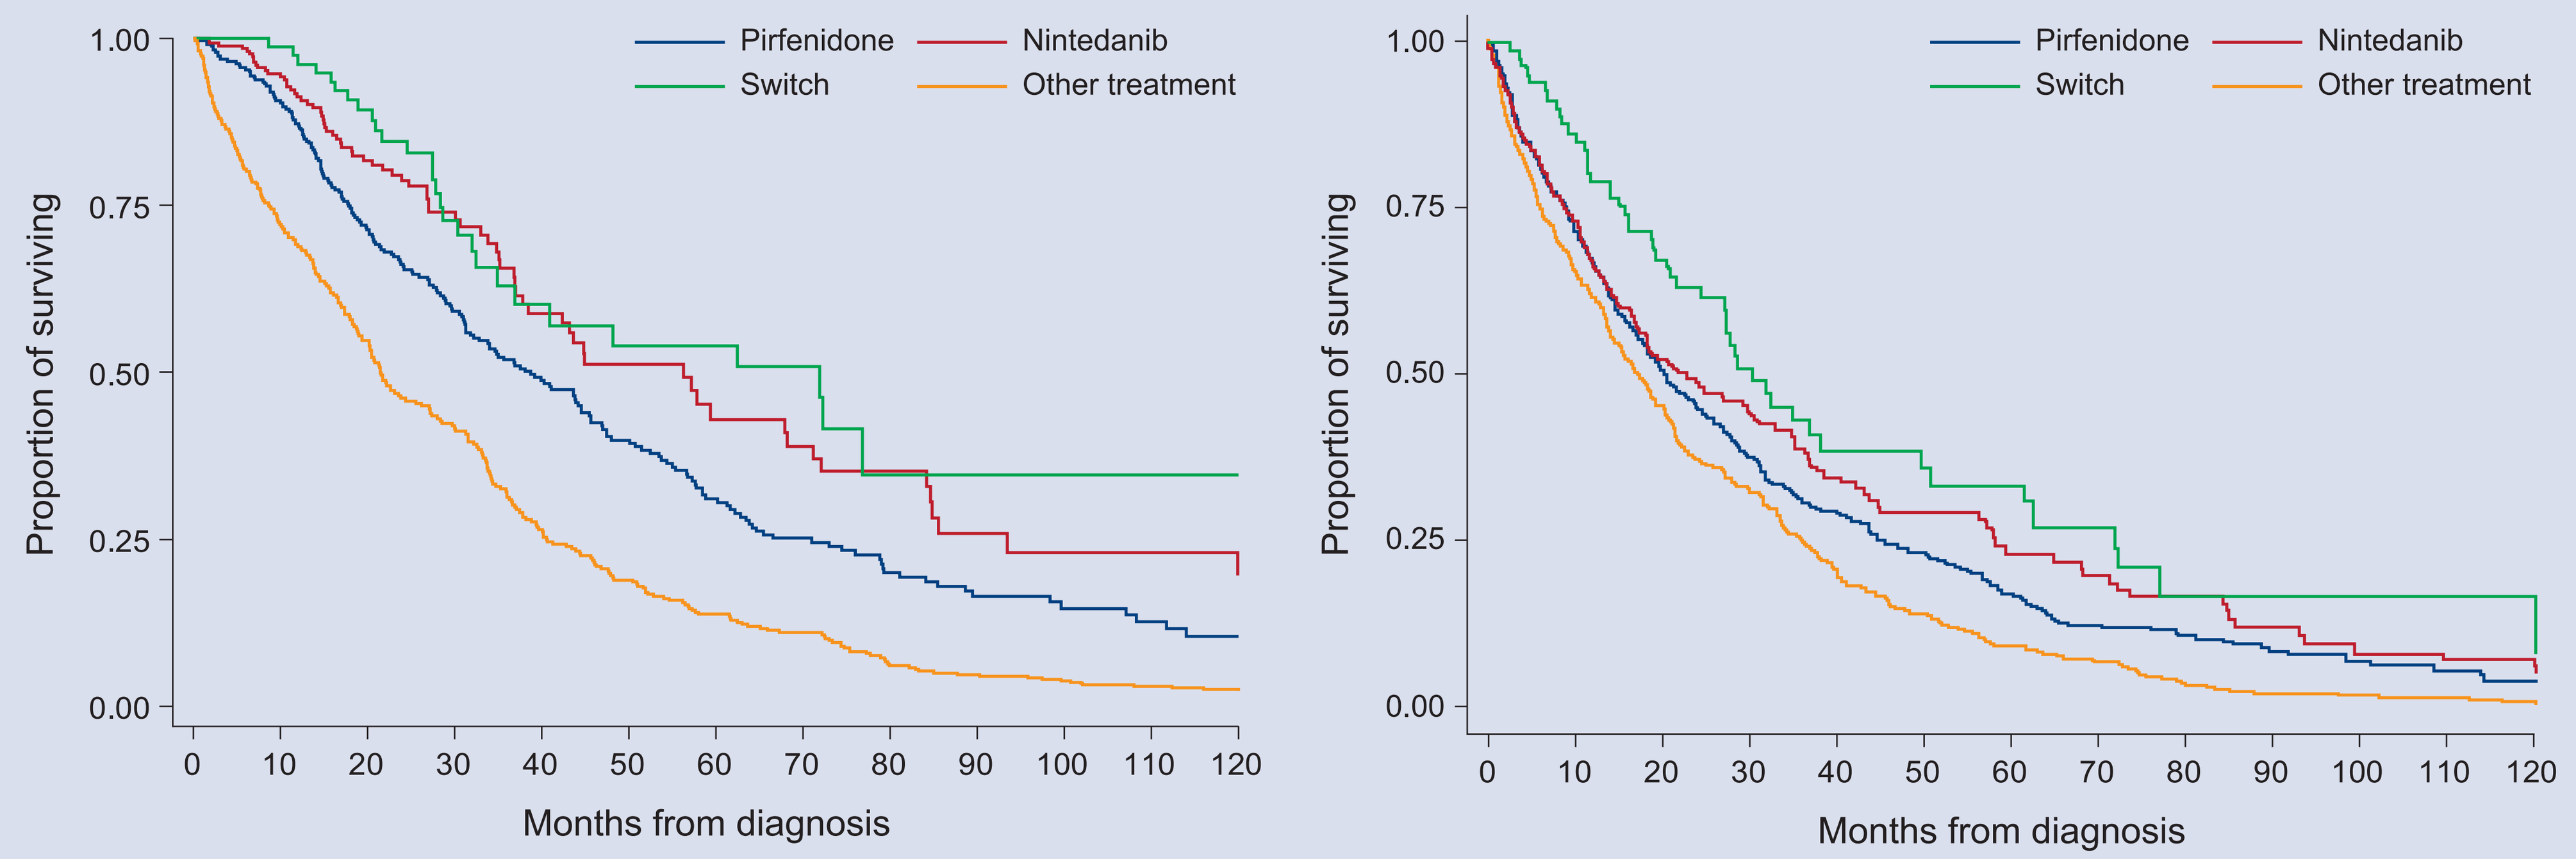
Pirfenidone
Nintedanib
Switch
Other treatment
Proportion of surviving
1.00
0.75
0.50
0.25
0.00
0
10
20
30
40
50
60
70
80
90
100
110
120
Months from diagnosis

	Median survival (months)	1-year survival (95% CI)	5-year survival (95% CI)	10-year survival (95% CI)	Median survival (months)	1-year survival (95% CI)	5-year survival (95% CI)	10-year survival (95% CI)
Pirfenidone	38.7	0.872 (0.831–0.904)	0.311 (0.251–0.372)	0.105 (0.060–0.163)	20.5	0.673 (0.621–0.719)	0.173 (0.133–0.217)	0.040 (0.018–0.076)
Nintedanib	56.3	0.912 (0.867–0.943)	0.430 (0.314–0.540)	0.197 (0.097–0.323)	22.9	0.669 (0.609–0.723)	0.232 (0.162–0.310)	0.055 (0.021–0.112)
Switch	71.9	0.961 (0.884–0.987)	0.540 (0.382–0.674)	0.347 (0.171–0.530)	30.4	0.789 (0.683–0.863)	0.333 (0.210–0.460)	0.084 (0.010–0.268)
Other treatment	21.4	0.688 (0.633–0.737)	0.138 (0.101–0.180)	0.023 (0.011–0.042)	17.3	0.617 (0.561–0.669)	0.094 (0.066–0.129)	0.008 (0.003–0.020)

[a]Long-term survival according to TL_{CO} % at diagnosis. [b]Progression defined as death, transplant or progression (TL_{CO} or FVC decline).

Figure 2. Survival in patients who switched treatment due to AEs or lack of treatment effect

A. OS **B. PFS**

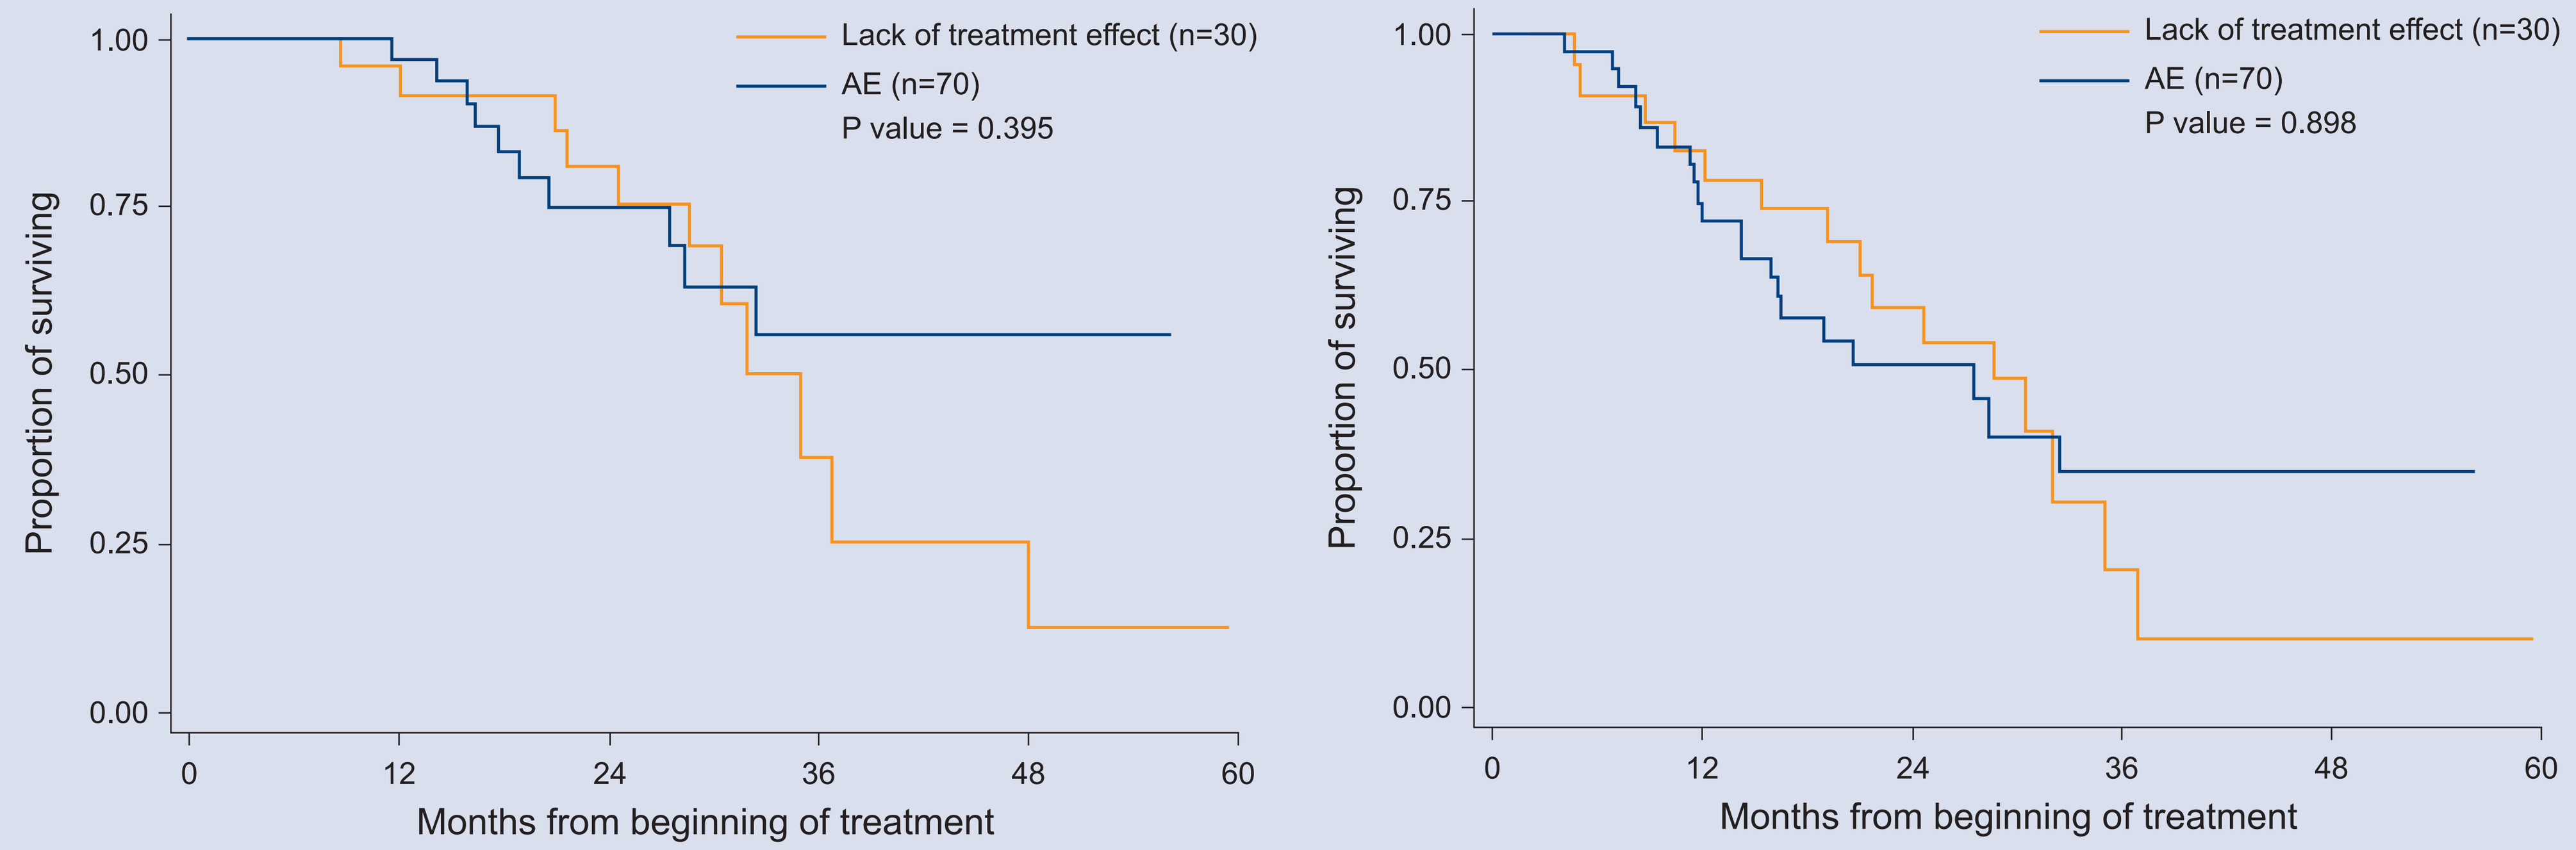
Lack of treatment effect (n=30)
AE (n=70)
P value = 0.395
P value = 0.898
Proportion of surviving
1.00
0.75
0.50
0.25
0.00
0
12
24
36
48
60
Months from beginning of treatment

	Median survival (months)	1-year survival (95% CI)	2-year survival (95% CI)	5-year survival (95% CI)	Median survival (months)	1-year survival (95% CI)	2-year survival (95% CI)	5-year survival (95% CI)
AE	76.9	0.969 (0.798–0.996)	0.752 (0.546–0.874)	0.561 (0.316–0.748)	27.4	0.722 (0.544–0.840)	0.509 (0.331–0.662)	0.351 (0.173–0.535)
Lack of treatment effect	34.9	0.915 (0.700–0.978)	0.810 (0.566–0.925)	0.126 (0.008–0.417)	28.6	0.783 (0.555–0.903)	0.592 (0.359–0.764)	0.102 (0.007–0.352)

[a]Long-term survival according to TL_{CO} % at diagnosis. [b]Progression defined as death, transplant or progression (TL_{CO} or FVC decline).

Reasons for Discontinuing Therapy

- Patients discontinued their first therapy due to AEs (47.3%) or lack of treatment effect (14.9%).
- Fifty patients discontinued pirfenidone and 20 discontinued nintedanib as first therapy due to AEs (Table 2).
- In patients who switched treatment, the most common AEs leading to discontinuation of first therapy with pirfenidone were rash (16.9%), nausea (16.9%) and diarrhoea (10.2%).
- In patients treated with nintedanib, the most common AEs leading to discontinuation of first therapy were diarrhoea (41.7%), emesis (16.7%) and elevated liver enzymes (12.5%) (Table 3).

Table 2. AEs leading to discontinuation of first therapy

AE[a]	Pirfenidone (n=50)	Nintedanib (n=20)
Diarrhoea	6 (10.2)	10 (41.7)
Skin problems, rash	10 (16.9)	1 (4.2)
Nausea	10 (16.9)	0
Emesis	5 (8.5)	4 (16.7)
Elevated liver enzymes	4 (6.8)	3 (12.5)
Photosensitivity/-toxicity	6 (10.2)	0
Weight loss	5 (8.5)	0
Abdominal pain	1 (1.7)	2 (8.3)
Joint pain	3 (5.1)	0
Vertigo	2 (3.4)	0
Bleeding	0	2 (8.3)
Fatigue	2 (3.4)	0
Hypersensitive reaction	1 (1.7)	0
Hypertension	0	1 (4.2)
Other	4 (6.8)	1 (4.2)

Patients may have experienced more than one AE and provided multiple reasons for therapy discontinuation. [a]Data are n (%).

Conclusions

- As in all retrospective studies, these results are unadjusted and do not account for potential differences between treatment groups.
- Results show a consistent beneficial effect of antifibrotic treatment on OS and PFS in a large cohort of real-world patients with IPF.
- Median OS was significantly longer in patients who received nintedanib (vs pirfenidone) and in those who switched treatment (vs other treatment).
- Switching antifibrotic treatments was not detrimental and can benefit some patients.

References

1. Richeldi L, et al. N Engl J Med 2014; 370(22):2071–2082;
2. Richeldi L. Thorax 2018; 73(6):581–583;
3. King TE Jr, et al. N Engl J Med 2014; 370(22):2083–2092;
4. European Medicines Agency. OFEV® (nintedanib) Summary of Product Characteristics. 2018;
5. US Food and Drug Administration. OFEV® (nintedanib) prescribing information. 2014. Available here: https://www.accessdata.fda;
6. Genentech. ESBRIET® (pirfenidone) prescribing information. 2017;
7. European Medicines Agency. Pirfenidone Summary of Product Characteristics. 2017.

Disclosures

MV received fees from Boehringer Ingelheim and Roche for consultancy, advisory boards and presentations, and an independent grant from Roche; MSte received fees for advisory boards from Boehringer Ingelheim, Roche and Teva; NM received fees for advisory boards and presentations from Boehringer Ingelheim and Roche; KL received payments for lectures, advisory boards, research and congress participation from Boehringer Ingelheim and Roche; DJ was an advisory board member and received author fees from Boehringer Ingelheim and Roche; JTT was an advisory board member and received fees for lectures from Boehringer Ingelheim and Roche; MStu received research grants from Boehringer Ingelheim and Roche; NS was an advisory board member for Boehringer Ingelheim; VM, MK, SL, KH and LD declare no conflicts of interest.

Acknowledgements

This study was supported by Boehringer Ingelheim. The EMPIRE registry is financially supported by Boehringer Ingelheim and Roche. Helen Keyworth at Nucleus Global provided editorial assistance in the development of this poster, funded by Boehringer Ingelheim. The authors would like to thank all the EMPIRE registry patients, and they also extend their thanks to the investigators.

Abbreviations

MV received fees from Boehringer Ingelheim and Roche for consultancy, advisory boards and presentations, and an independent grant from Roche; AE, adverse event; ANOVA, analysis of variance; AEATS, American Thoracic Society; CI, confidence interval; EMPIRE, European Multipartner IPF registry; ERS, European Respiratory Society; FVC, forced vital capacity; HRCT, high-resolution computerised tomography; IPF, interstitial pulmonary fibrosis; OS, overall survival; PFS, progression-free survival; TL_{CO}, transfer factor for carbon monoxide; UIP, usual interstitial pneumonia.

1. LÉKAŘSKÁ FAKULTA Univerzita Karlova · Thomayerova nemocnice · MUNI MED · MU IBA · EMPIRE (IPF)

Poster presented at the European Respiratory Society Annual Meeting, 28 September – 2 October 2019, Madrid, Spain.